This poster (P-85) appeared at the 88th Annual Meeting of the British Association of Dermatologists, July 2008
[DB gel refers to Doublebase Gel (Dermal Laboratories)]

An *in vivo* comparison of the cumulative effect on skin hydration of two topically applied formulations, DB gel and Aqueous Cream

J.Gallagher[1], P. Rosher[1], J. Walker[1], V.A. Hart[2]
[1]Dermal Laboratories Ltd, Hitchin, UK, [2]RSSL, Reading, UK

Atopic eczema

Atopic eczema is the most common form of eczema, which usually develops in early childhood. It is characterised by chronic itchy and inflammatory skin, often involving the flexures[1]. Recent NICE guidance on atopic eczema management in children from birth to the age of 12 years[2] establishes emollient therapy as the treatment modality that should underpin all else. The guidance recommends continuing emollient therapy even when the skin is clear, and healthcare professionals are advised to offer children with the condition a regime of complete emollient therapy involving a choice of non perfumed emollients to use every day for moisturising, washing and bathing. Although originally devised as a soap substitute, Aqueous Cream BP attracts prescriptions for continuous applied use, owing to its relatively low cost. The hydration properties of Aqueous Cream are, however, largely assumed rather than proven. In addition, the cutaneous reactions to Aqueous Cream in children with atopic eczema have been well documented[3].

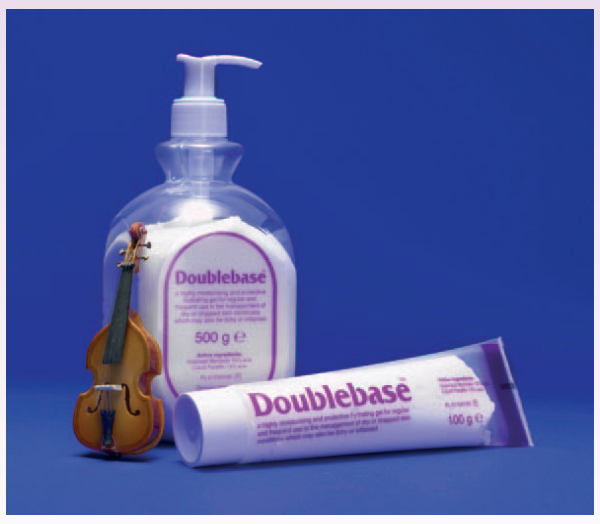
Doublebase
500 g ℮
Doublebase
100 g ℮

The aim of this clinical study conducted with full ethics committee approval and in accordance with GCP requirements was to compare the cumulative effects on skin hydration of Aqueous Cream ("AC") with that of a proprietary emollient, Doublebase Gel ("DB"), which, unlike Aqueous Cream, is specially formulated for continuous use. Both preparations contain the same concentration of oily ingredients (30%w/w).

Methodology

- The study was an assessor-blind bilateral comparison in twenty adult females with self perceived dry skin on their lower legs.
- Written informed consents were obtained and witnessed on day 1. Exclusion criteria were: significant concurrent illness or skin disease; history of allergy relevant to the test products or their ingredients; use of any topical or systemic treatment likely to affect skin response; visible skin abnormality or excessive hair growth, irritation, tattoos, scars or birthmarks at the test measurement sites; participation in any other study presently or within the past 3 months; breastfeeding and pregnancy (actually or potentially).
- Baseline corneometric measurements of skin hydration at sites on both lower legs were performed on day 1. Subjects were then given the two test products, presented in identical 500g bottles fitted with metered pump dispensers. The two products were randomly assigned for application to each subject's right and left lower legs in equal numbers. On days 1, 3 and 5, subjects were instructed to apply a fixed amount of each product (one actuation delivering 1.2ml) to their lower legs at approximately 09:00 hrs, after the first assessments when at the test centre, and again, at home, at approximately 12 noon and again at 17:00 hrs. They attended the centre approximately 2 hours after the third application on these days for the skin hydration measurements to be repeated. On intervening days 2, 4, 6 and 7, the subjects were instructed to re-apply the products to each leg at approximately 09:00hrs, midday and 17:00 hours. There were no test centre visits or corneometry measurements on these days. Subjects then attended the centre on the morning of day 8 for a final skin hydration measurement.
- Corneometry measurements were performed using the Multiprobe Adapter MPA5 with Corneometer CM825 probe (Hydration) (ex Courage-Khazaka electronic, Germany). Measurements were performed in triplicate to the same skin areas, located by a template for each subject. The data were analysed using Wilcoxon's rank-sum test with PROC NPAR1WAY.
- The per protocol primary efficacy analysis was the improvement in skin moisturisation measured by corneometry.

Results & Discussion

The mean corneometry measurements, and their mean differences from baseline, for each product are summarised in Figures 1 and 2.

All of the mean differences from baseline were several fold higher for DB gel than for AC, and the differences in favour of DB were statistically significant (p≤0.05) with the exception of the morning measurement on day 3.

Figure 1. Mean Corneometer Readings

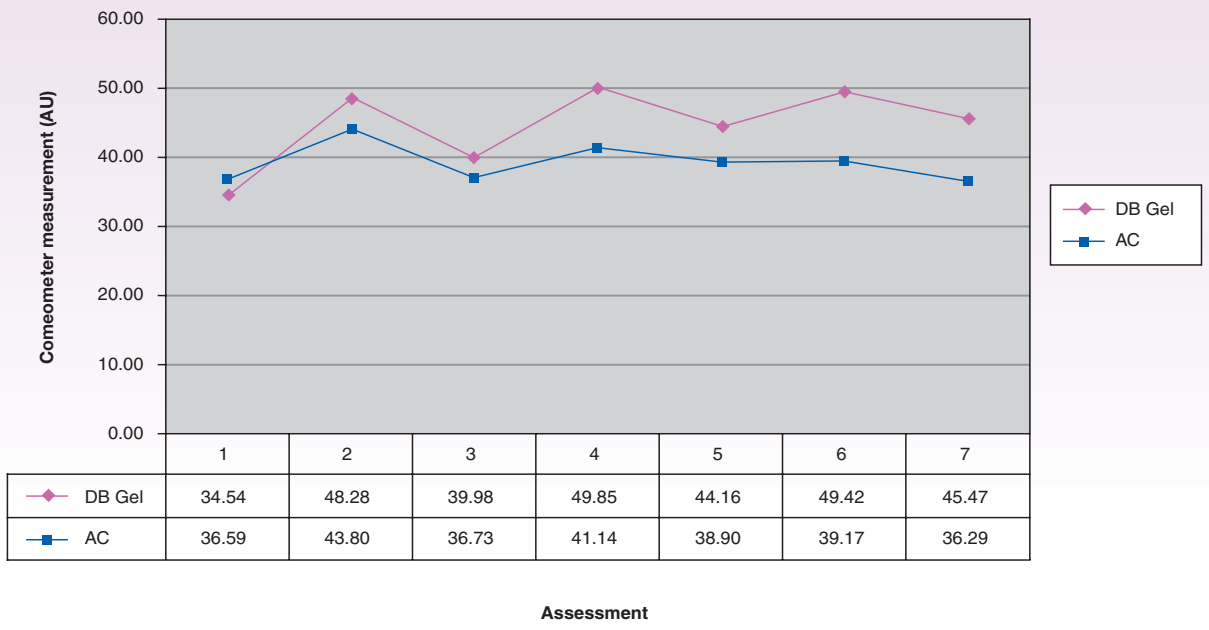
Corneometer measurement (AU)
60.00
50.00
40.00
30.00
20.00
10.00
0.00
DB Gel
AC
Assessment

	1	2	3	4	5	6	7
DB Gel	34.54	48.28	39.98	49.85	44.16	49.42	45.47
AC	36.59	43.80	36.73	41.14	38.90	39.17	36.29

Corneometer readings taken immediately before the first application of the day (assessments 3, 5 & 7) were lower than the maximum reached the previous evening (assessments 2, 4 & 6). This is a consequence of the 14 hour or so overnight period with no re-application, and subjects washing, bathing or showering in the interim.

Figure 2. Mean Difference from Baseline Corneometer Readings

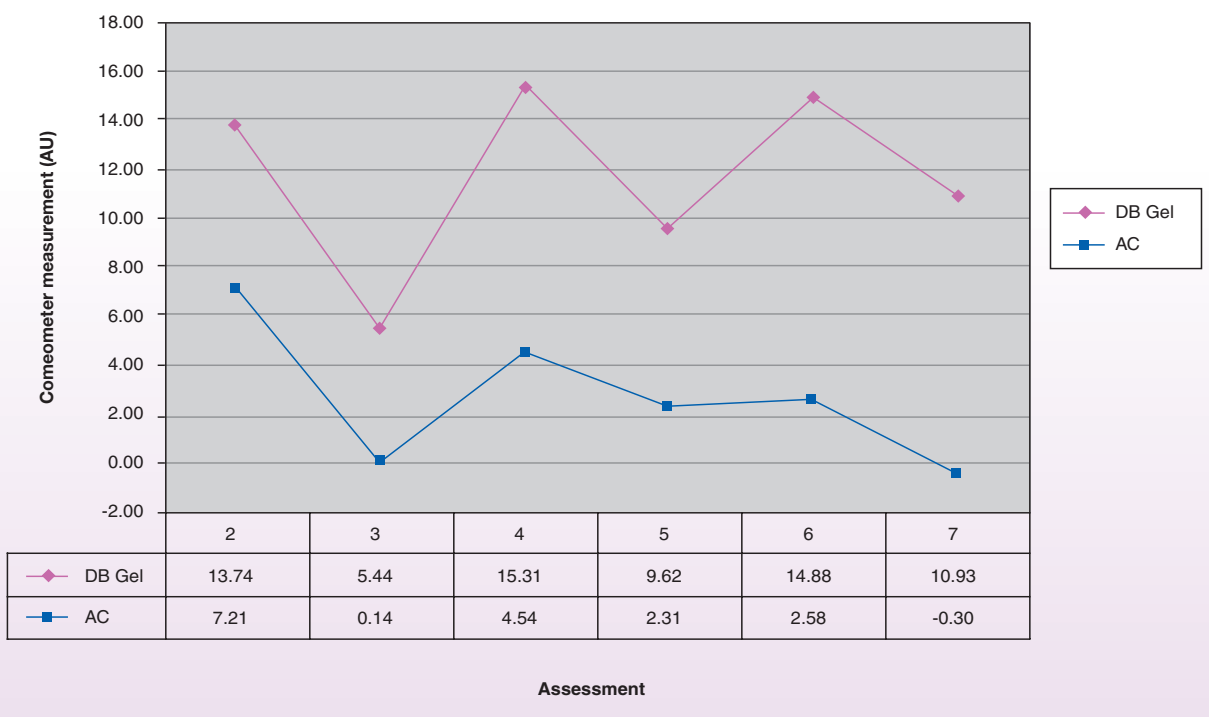
Corneometer measurement (AU)
18.00
16.00
14.00
12.00
10.00
8.00
6.00
4.00
2.00
0.00
-2.00
DB Gel
AC
Assessment

	2	3	4	5	6	7
DB Gel	13.74	5.44	15.31	9.62	14.88	10.93
AC	7.21	0.14	4.54	2.31	2.58	-0.30

Notably, however, whereas corneometer readings for AC returned to baseline overnight, the DB gel readings did not, and showed a stepwise, cumulative increase over time (Figure 2).
After 7 days' continuous use skin hydration levels with AC were barely improved, whereas the values for DB sites were elevated by more than 10 units (the amount generally being regarded as being therapeutically relevant).

Conclusion

The treatment of atopic eczema in children requires continuous use of emollient topical preparations in order to maintain skin hydration. Although it is a common belief among healthcare professionals that any topical preparation possesses good hydration properties, we have demonstrated that one frequently prescribed emollient, Aqueous Cream BP, does not improve the skin hydration levels even after 7 days of continuous use. In contrast, Doublebase Gel has significant, and cumulative beneficial effect on skin hydration.

References: **1-** Loden M The Skin Barrier and Use of Moisturizers in Atopic Dermatitis, *Clinics in Dermatology*, 2003;**21**:145-157. **2-** NICE. Atopic eczema in children: management of atopic eczema in children from birth up to the age of 12 years. CG57. December 2007 (www.nice.org.uk/CG057). **3-** Cork MJ, Holden C, Carr J, Berry V, Tazi-Abnini R and Ward SJ, An audit of adverse drug reactions to aqueous cream in children with atopic eczema, *The Pharmaceutical Journal*, 2003:271:747-748.

Please turn over for poster summary

Choosing a Suitable Leave-on Emollient.

Aqueous cream incorporating sodium lauryl sulphate was designed as a soap substitute. In spite of this it is often prescribed as a leave on emollient. However, there are well reported irritancy or 'stinging' problems associated with aqueous cream[3] when used as a leave-on moisturiser.

"Cutaneous reactions to aqueous cream in children with atopic eczema are so common that it should only be used as a soap substitute and not as a "leave on" emollient."[3]

The trial summarised overleaf, shows that Doublebase exhibits significantly improved hydration properties compared to aqueous cream.

"…..Aqueous Cream BP, does not improve the skin hydration levels even after 7 days of continuous use. In contrast, Doublebase Gel has significant, and cumulative beneficial effect on skin hydration."

Summary of Poster Overleaf:

- The study was an assessor-blind bilateral comparison in twenty adult females, with self perceived dry skin on their lower legs.
- The two test products were presented in identical 500g bottles fitted with metered pump dispensers.
- One actuation delivering 1.2ml of Doublebase Gel or Aqueous Cream was applied to the right or left lower leg respectively, three times a day for seven days. These were applied at 09:00 hrs, midday and 17:00 hrs.
- Two corneometer readings (to measure skin hydration) were taken on days 1, 3 and 5. Readings were taken prior to the first application of the day and 2 hours after the last application of the day. A final reading was taken on the morning of day 8.

Conclusion:

The treatment of atopic eczema in children requires continuous use of emollient topical preparations in order to maintain skin hydration. Although it is a common belief among healthcare professionals that any topical preparation possesses good hydration properties, we have demonstrated that one frequently prescribed emollient, Aqueous Cream BP, does not improve the skin hydration levels even after 7 days of continuous use. In contrast, Doublebase Gel has significant, and cumulative beneficial effect on skin hydration.

Doublebase™ Gel

Isopropyl myristate 15% w/w, liquid paraffin 15% w/w.

Adverse events should be reported. Reporting forms and information can be found at yellowcard.mhra.gov.uk
Adverse events should also be reported to Dermal.

Click **here** for the Doublebase Range Prescribing Information or scan the QR code below

DERMAL
TOPICAL INNOVATION
www.dermal.co.uk

Further information is available from:
Dermal Laboratories Limited,
Tatmore Place, Gosmore, Hitchin,
Herts SG4 7QR

DXXPR399/JAN25