Data presented at the Annual Meeting of the Austrian Society of Dermatology and Venereology (ÖGDV), November 2024, Graz, Austria.

Evaluation of a nicotinamide-containing emollient for moderate atopic eczema in paediatric patients: A prospective, multi-centre GP study reflecting real-life settings

Jasmina Gallagher, Alastair Cruickshank, Jennine Walker, Valerie Hart
Research and Development, Dermal Laboratories Ltd., Hitchin, UK

Introduction

Moderate atopic eczema is characterised by recurrent inflammation and itching, affecting the patient's quality of life; this condition is frequently difficult to control and treat. There is an increasing need for treatments, such as emollients containing additional anti-inflammatory therapeutic properties, due to the limitations of topical corticosteroids.

Methodology

This was a prospective, open-label study conducted in 11 GP centres across the UK, involving 60 screened children (aged 1 year to 15 years) with moderate atopic eczema. The test product was Adex Gel (Dermal Laboratories Ltd., UK), an emollient containing an ancillary anti-inflammatory medicinal substance, nicotinamide. Adex Gel was used three times daily for 4 weeks instead of usual emollient or as the first-line treatment for moderate atopic eczema without supplementary use of any oral or topical steroids or immunomodulators. The clinical endpoints were the change from baseline in Scoring Atopic Dermatitis (SCORAD)* measurements after 2 and 4 weeks of treatment and the change from baseline in Children's Dermatology Life Quality Index (CDLQI)** questionnaire after 4 weeks of treatment.

Results

The mean disease severity score (SCORAD) improved significantly from 37.14 (moderate atopic eczema) at baseline, to 22.56 (mild atopic eczema) after 2 weeks and to 18.48 (mild atopic eczema) after 4 weeks, per protocol analysis of 41 children.

Figure 1 - Per Protocol Analysis - using just the patients with data at each visit

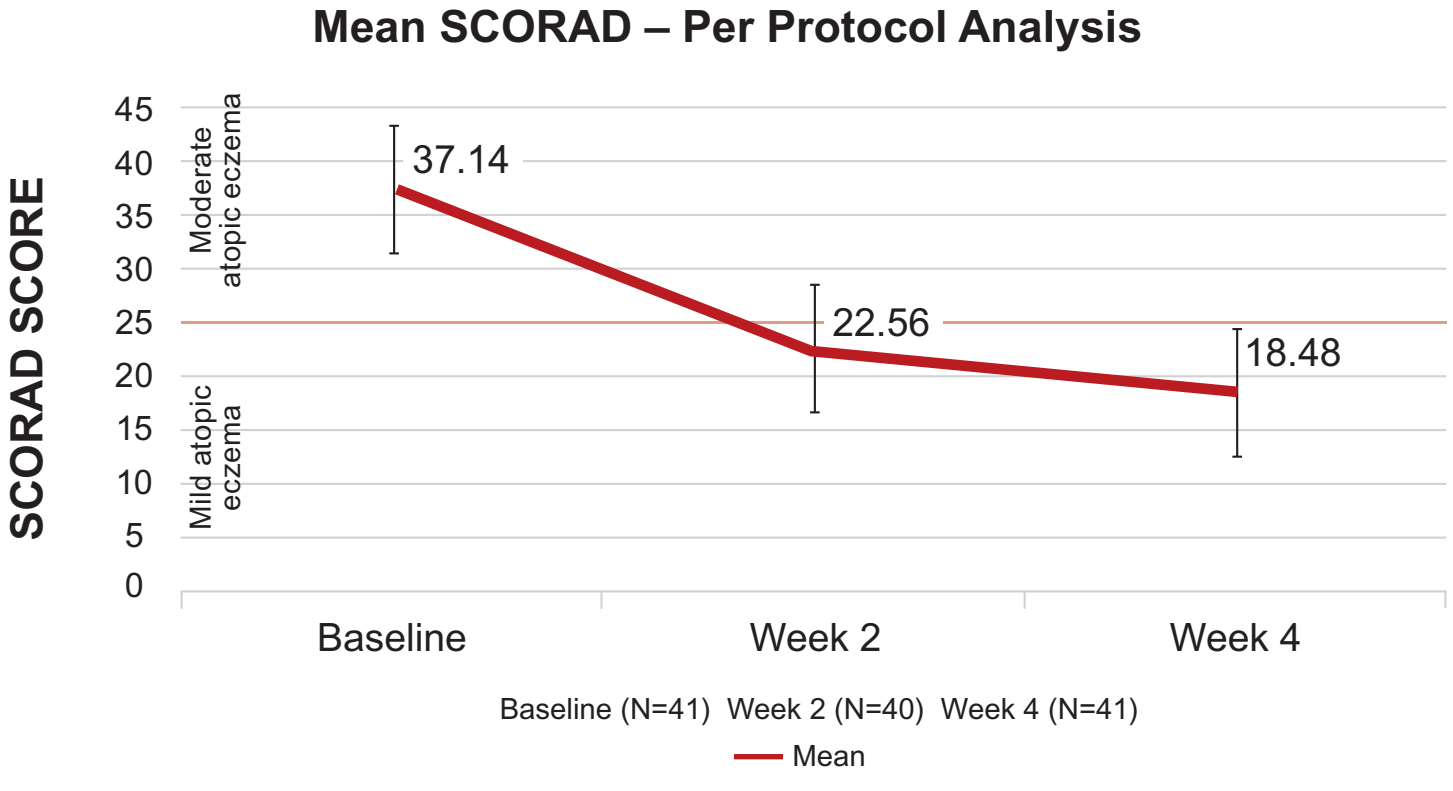
Mean SCORAD – Per Protocol Analysis
SCORAD SCORE
45
40
35
30
25
20
15
10
5
0
Moderate atopic eczema
Mild atopic eczema
37.14
22.56
18.48
Baseline
Week 2
Week 4
Baseline (N=41) Week 2 (N=40) Week 4 (N=41)
Mean

There was a statistically significant improvement in SCORAD at both week 2 and week 4 (p<0.0001) in the per protocol analysis (n=41). An improvement in SCORAD of 14.45 after 2 weeks and 18.67 after 4 weeks treatment was observed.

Table 1 – SCORAD per protocol analysis

	SCORAD Change from baseline	95% CI	*p*-value
Week 2 (N=41)	-14.45	-17.64, -11.27	<0.0001
Week 4 (N=41)	-18.67	-21.99, -15.34	<0.0001

	% Responders Minimal Clinically Important Difference (MCID)*	95% CI
Week 2 (N=41)	75.6%	60.7, 86.2%
Week 4 (N=41)	85.4%	71.6, 93.1%

Missing values imputed using baseline observation carried forward

The total CDLQI score improved from 9.3 at baseline to 3.7 at week 4 showing a statistically significant improvement of 5.6 (p<0.0001).

Table 2 – Responder analysis for CDLQI – per protocol population

	Responders meeting Minimal Clinically Important Difference (MCID)**	95% CI
Week 4 (N=41)	56.1%	41.0, 70.1%

Missing values imputed using non-responder imputation

Related adverse events were reported, with stinging, itching, redness and worsening of the eczema symptoms being the most common, but these are expected reactions with any emollient.

Conclusion

Adex Gel has been proven to be an effective treatment for children with moderate eczema, as confirmed through this assessment in NHS GP settings, avoiding the need for escalation to topical corticosteroids.

This evidence underscores the value of Adex Gel in improving clinical outcomes for children with moderate atopic eczema in a real-world setting, demonstrating both its efficacy and safety in everyday use.

*SCORAD is a tool used in clinical trials to assess atopic dermatitis severity based on disease area, intensity and subjective symptoms (itch and sleeplessness). SCORAD-Index = Mild 0-24; Moderate 25-50; Severe 51-100 (Willemsen, M.G., *et al*., Determining the severity of atopic dermatitis in children presenting in general practice: an easy and fast method. *Dermatol Res Pract*, ***2009**: 357046*).
Improved SCORAD outcomes of within-patient change of 8.7 is considered the Minimal Clinically Important Difference (MCID) or above.
**CDLQI is designed to measure the impact of any skin disease on the lives of children. For the CDLQI the within-patient change of 6 is considered the MCID.

701600 ADX140/B/NOV24